IMPACT OF GREXIT ON PHARMACEUTICAL PRICING: AN INTERNATIONAL REFERENCE PRICING ANALYSIS

Jeffrey Weiss[1], Parvez Hakim[1], Rikki Sawhney[2], Johan Kjeldgaard-Pedersen[3], Ravi Degun[1], Ken Walsh[4]

[1]Navigant, London, United Kingdom; [2]GSK Knowledge Centre, Haryana, India; [3]data2impact, Copenhagen, Denmark; [4]GlaxoSmithKline, London, United Kingdom

INTRODUCTION

- The possibility of Greece leaving the Eurozone is well-publicised, but its impact on the pharmaceutical industry has not been fully determined
- The majority of established pharmaceutical markets use pricing rules that reference products both across IRP (International Reference Pricing) and within TRP (Therapeutic Reference Pricing) country-lines, as effective measures to control the price of pharmaceutical products
- There is a growing need to understand how Grexit will impact pricing mechanisms through direct or indirect means, including changes that could be explored by a national ministry of health
- Parallel trade may become a significant issue with neighbouring markets attempting to secure cheaper sources of medicine, leading to shortages in the market potentially impacting patient access

METHODOLOGY

PRICES BY MARKETS WERE OBTAINED USING AGGREGATED SALES AND VOLUME DATA ACROSS RESPIRATORY PRODUCTS USING IMS MIDAS, APPLYING A WEIGHTED AVERAGE

Weighted Average PTW Price of Respiratory Products by Market at Month 1

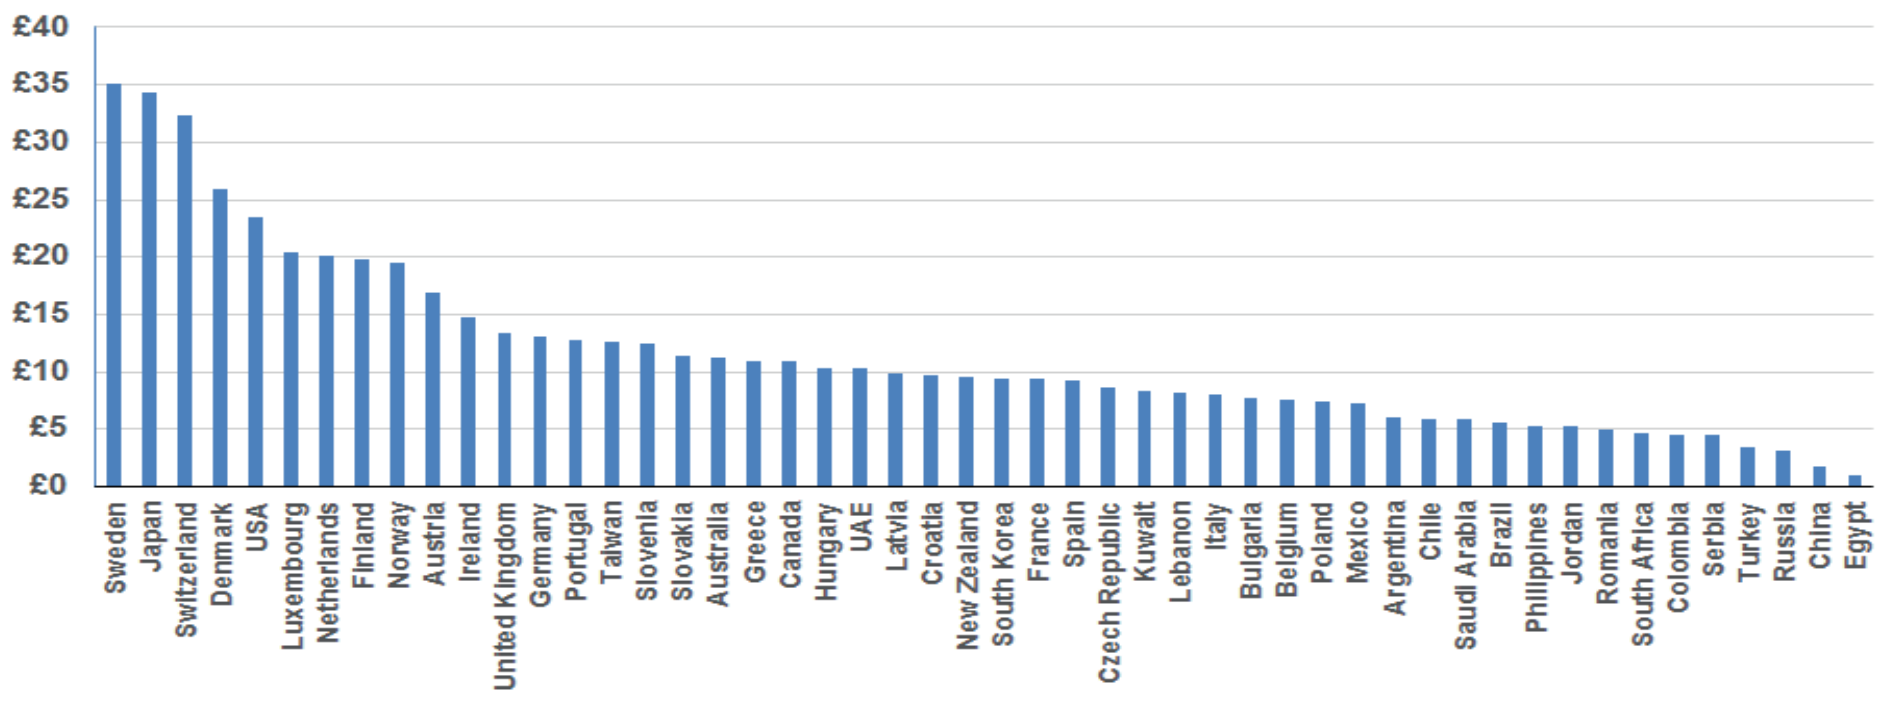

£40
£35
£30
£25
£20
£15
£10
£5
£0
Sweden
Japan
Switzerland
Denmark
USA
Luxembourg
Netherlands
Finland
Norway
Austria
Ireland
United Kingdom
Germany
Portugal
Taiwan
Slovenia
Slovakia
Australia
Greece
Canada
Hungary
UAE
Latvia
Croatia
New Zealand
South Korea
France
Spain
Czech Republic
Kuwait
Lebanon
Italy
Bulgaria
Belgium
Poland
Mexico
Argentina
Chile
Saudi Arabia
Brazil
Philippines
Jordan
Romania
South Africa
Colombia
Serbia
Turkey
Russia
China
Egypt

1. Navigant's PROFILE model (Pricing and Revenue Optimisation for International Launch Excellence), was used to simulate the impact of IRP and TRP, as well as parallel trade and generic entry, to quantitatively assess the impact on drug prices across 50 markets throughout Europe, North America, Latin America, Middle East and Asia
2. Respiratory products were analysed as a class to prevent a biased analysis towards particular manufacturers, assessing three potential scenarios with respect to Grexit:
 - Currency deflation of 50% in Greece resulting from a shift to the Greek Drachma
 - Artificially freezing the visible price in Greece in Euros
 - Markets pursuing equitable alternatives to Greece by updating their reference baskets with Slovakia, a replacement low-priced EU market with similar referencing rules
3. Navigant's PROFILE model analysed relative changes in revenue due to IRP with and without parallel trade; volumes were aggregated by market and flat-lined across 10 years
4. In each scenario, changes were assumed after 18 months of baseline price evolution prior to IRP-specific price equilibrium being reached in Greece. These scenarios did not test for the implications of TRP and generic entry

RESULTS

- In Scenario 1 (Figure 1), currency deflation in Greece (Figure A) causes a cumulative reduction of 1.6% in overall revenue (Figure B). Spain is the greatest contributor, representing 26% of overall decline (38% decline within the market) due to referencing the minimum price in its basket
- In Scenario 2 (Figure 2), artificially freezing the Greek price in Euros (Figure A) has a negligible cumulative impact across all markets (Figure B), with Greece representing 98% of the additional gain vs. baseline by halting its semi-annual price re-referencing
- In Scenario 3 (Figure 3), when allowing for a price collapse in Greece in addition to basket substitution, an overall revenue decline of 0.1% is forecasted (Figure B), with Greece driving 100% of revenue loss
- Ignoring for Greece, basket substitution will have the greatest impact on Finland (73% contribution of overall revenue gain) by lessening the rate of its price decline
- Despite restricting market volumes parallel traded in the model, a Greek price decline still has a significant impact when compared with other scenarios (Figure C)

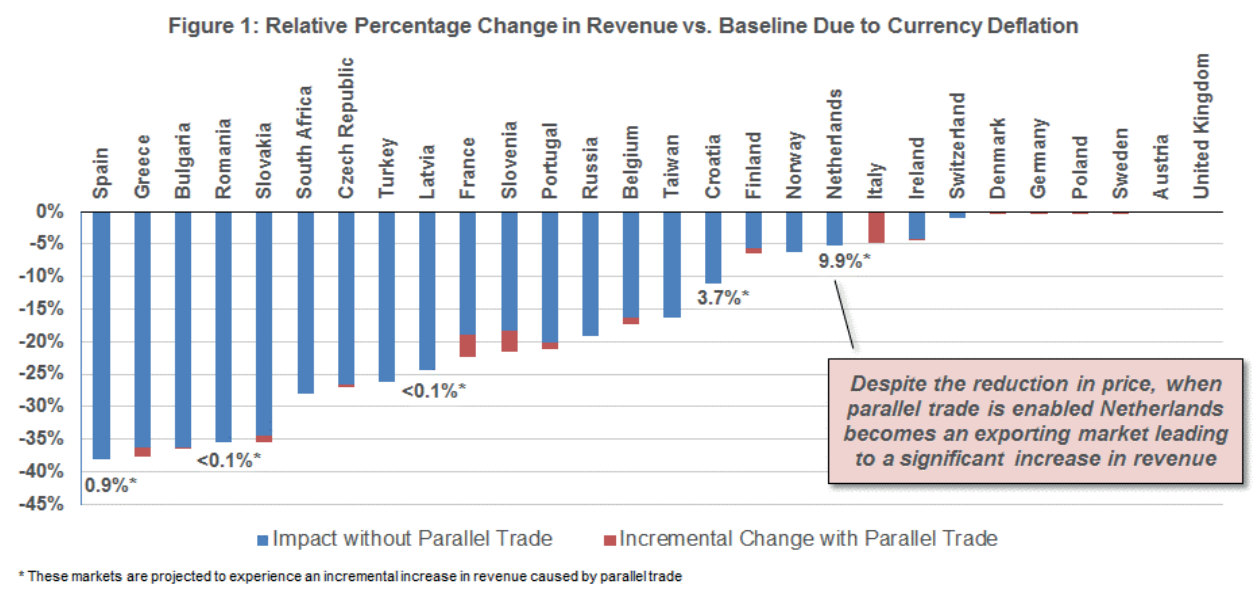

Spain
Greece
Bulgaria
Romania
Slovakia
South Africa
Czech Republic
Turkey
Latvia
France
Slovenia
Portugal
Russia
Belgium
Taiwan
Croatia
Finland
Norway
Netherlands
Italy
Ireland
Switzerland
Denmark
Germany
Poland
Sweden
Austria
United Kingdom
0%
-5%
-10%
-15%
-20%
-25%
-30%
-35%
-40%
-45%
0.9%*
<0.1%*
<0.1%*
3.7%*
9.9%*
Despite the reduction in price, when parallel trade is enabled Netherlands becomes an exporting market leading to a significant increase in revenue
Impact without Parallel Trade
Incremental Change with Parallel Trade
* These markets are projected to experience an incremental increase in revenue caused by parallel trade

Figure 1: Relative Percentage Change in Revenue vs. Baseline Due to Currency Deflation

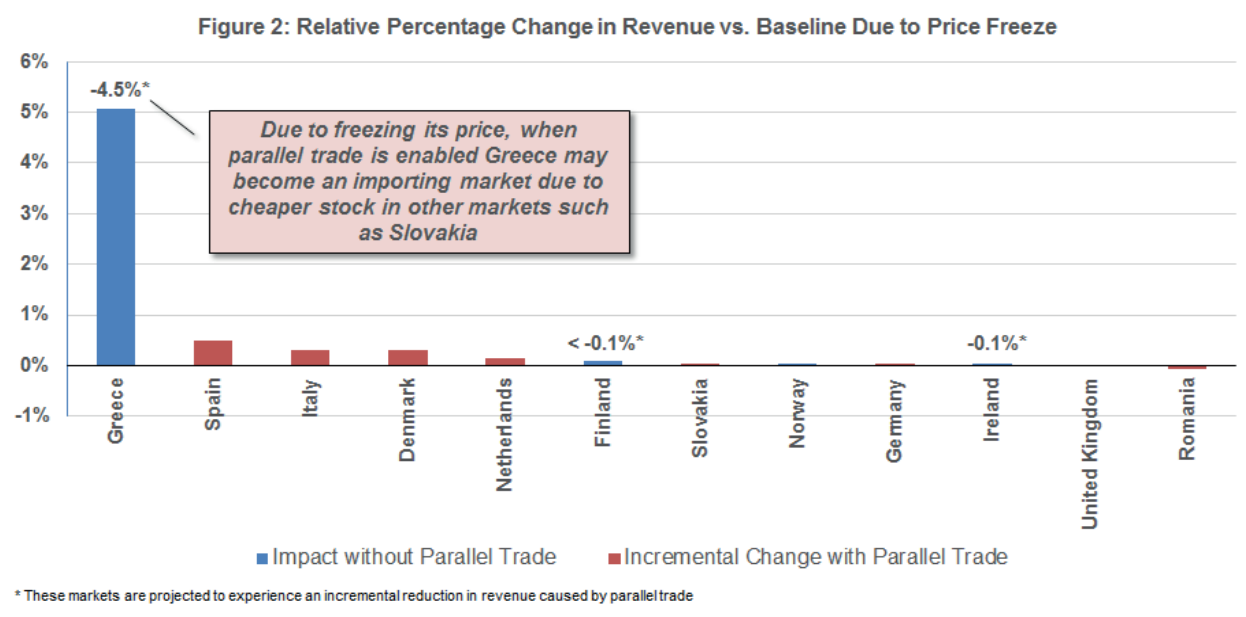

6%
5%
4%
3%
2%
1%
0%
-1%
-4.5%*
Due to freezing its price, when parallel trade is enabled Greece may become an importing market due to cheaper stock in other markets such as Slovakia
< -0.1%*
-0.1%*
Greece
Spain
Italy
Denmark
Netherlands
Finland
Slovakia
Norway
Germany
Ireland
United Kingdom
Romania
Impact without Parallel Trade
Incremental Change with Parallel Trade
* These markets are projected to experience an incremental reduction in revenue caused by parallel trade

Figure 2: Relative Percentage Change in Revenue vs. Baseline Due to Price Freeze

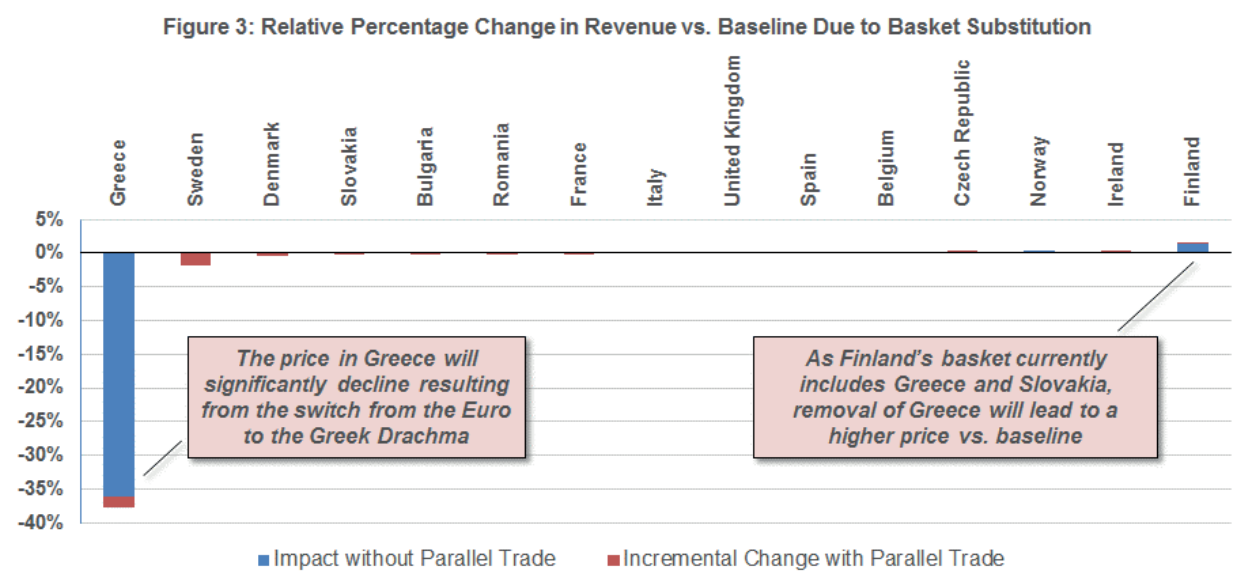

Greece
Sweden
Denmark
Slovakia
Bulgaria
Romania
France
Italy
United Kingdom
Spain
Belgium
Czech Republic
Norway
Ireland
Finland
5%
0%
-5%
-10%
-15%
-20%
-25%
-30%
-35%
-40%
The price in Greece will significantly decline resulting from the switch from the Euro to the Greek Drachma
As Finland's basket currently includes Greece and Slovakia, removal of Greece will lead to a higher price vs. baseline
Impact without Parallel Trade
Incremental Change with Parallel Trade

Figure 3: Relative Percentage Change in Revenue vs. Baseline Due to Basket Substitution

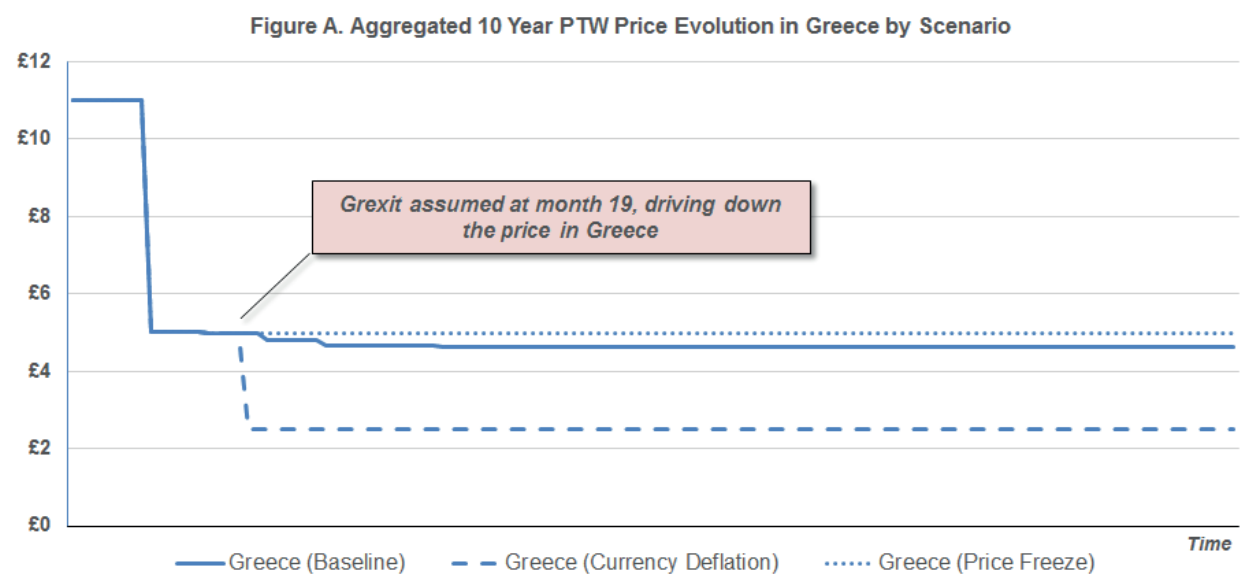

£12
£10
£8
£6
£4
£2
£0
Grexit assumed at month 19, driving down the price in Greece
Time
Greece (Baseline)
Greece (Currency Deflation)
Greece (Price Freeze)

Figure A. Aggregated 10 Year PTW Price Evolution in Greece by Scenario

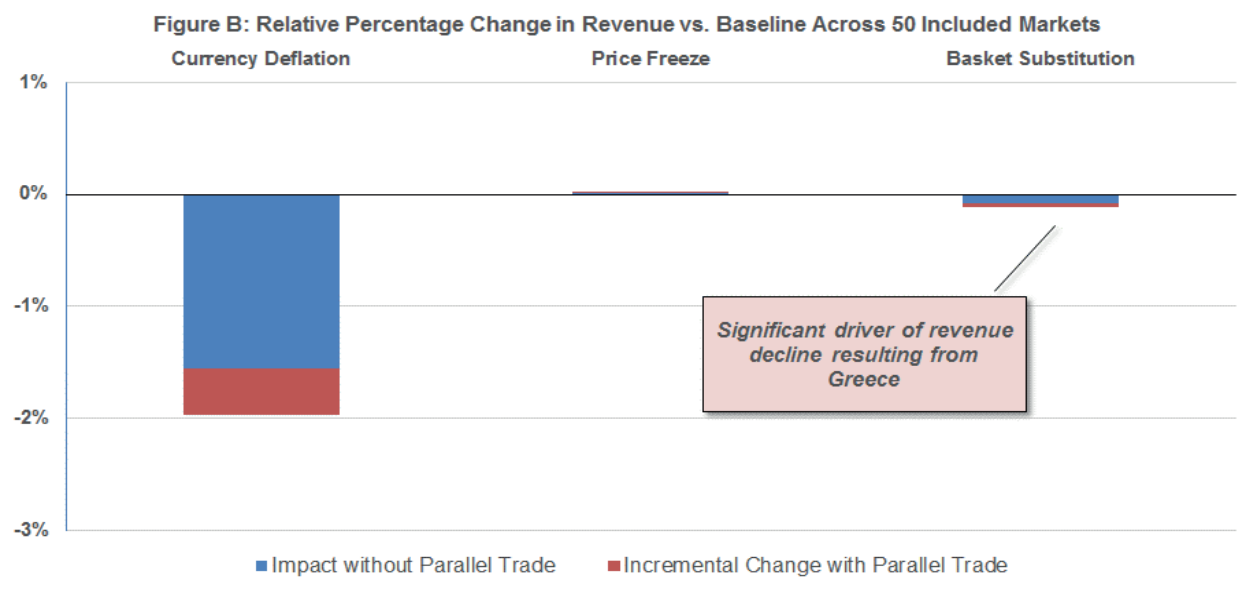

Currency Deflation
Price Freeze
Basket Substitution
1%
0%
-1%
-2%
-3%
Significant driver of revenue decline resulting from Greece
Impact without Parallel Trade
Incremental Change with Parallel Trade

Figure B: Relative Percentage Change in Revenue vs. Baseline Across 50 Included Markets

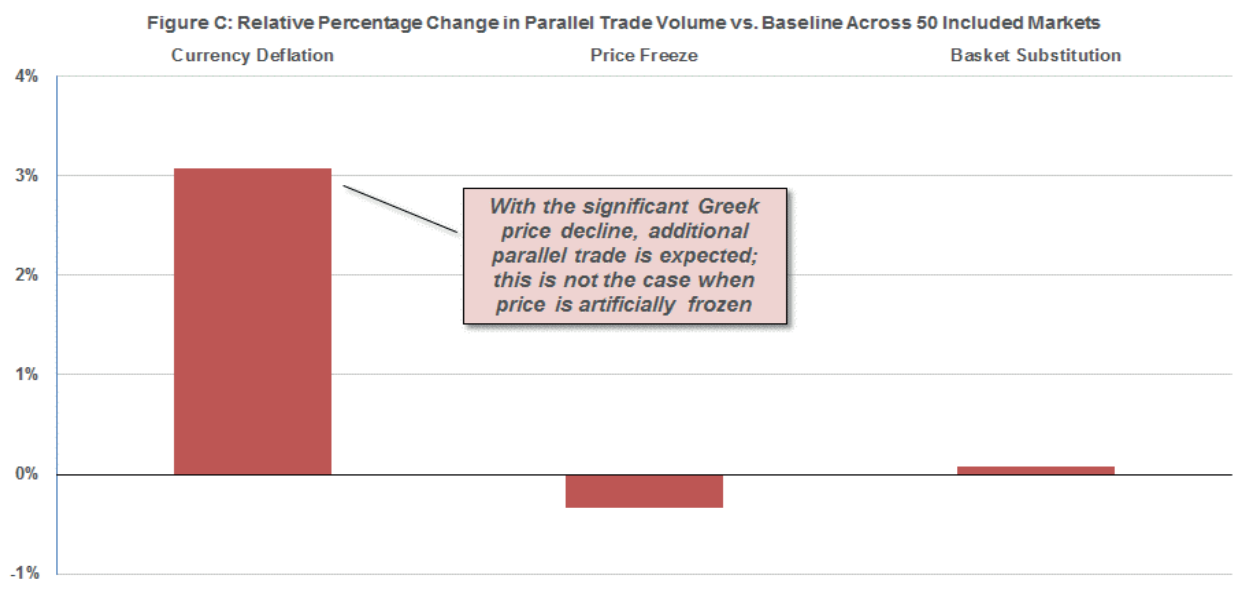

Currency Deflation
Price Freeze
Basket Substitution
4%
3%
2%
1%
0%
-1%
With the significant Greek price decline, additional parallel trade is expected; this is not the case when price is artificially frozen

Figure C: Relative Percentage Change in Parallel Trade Volume vs. Baseline Across 50 Included Markets

CONCLUSIONS

- The effects of Grexit go beyond Greece, as it highlights the importance for the industry to artificially stabilise the Greek price in Euros as a further preventative for future exchange rate fluctuations
- Assuming a constant price can be maintained - likely implemented through negotiation of an emerging confidential rebate after structure - coordination between ministries and manufactures to execute basket substitution may not be as critical
- Without a price freeze, legislation to limit parallel trade export may be required to prevent supply shortages of life saving medicines as well as considerable decline in global revenue
- An expansive pricing model employing varying IRP and TRP rules can be utilised to assist manufacturers in developing a comprehensive pricing strategy capable of evolving over time

Presented at: 2015 ISPOR 18th Annual European Congress, November 7-11, Milan, Italy
Sponsorship and data provided by GlaxoSmithKline with assistance from WNS

navigant.com

For more information on Navigant's Life Sciences practice,
please contact:

Susan Suponcic
Managing Director
E: susan.suponcic@navigant.com
T: +44(0)207 015 2336

Ravi Degun
Director
E: ravi.degun@navigant.com
T: +44(0)207 469 1125

navigant.com